P162

Comparative CNS Pharmacology of Tolebrutinib Versus Other BTK Inhibitor Candidates for Treating MS

Timothy J. Turner[1], Pricilla Brun[2], Dimitry Ofengheim[3], and Ross Gruber[3]

[1]Sanofi, Cambridge, MA, USA; [2]Sanofi, Montpelier, FR; [3]Sanofi, Framingham, MA, USA

OBJECTIVE

- **To evaluate the CNS pharmacological activity of 3 BTK inhibitors in Ph3 development in MS**

INTRODUCTION

- Bruton's Tyrosine Kinase (BTK) is a non-receptor tyrosine kinase expressed in myeloid cell types. BTK mediates signalling from certain cell surface receptors (e.g., BCR, FcγR, TLRs) to downstream elements crucial to immune cell function.
- Modulating BTK signalling within the CNS is a central tenet of our development program, we sought to characterize the relative ability of three candidate BTK inhibitors currently in Phase 3 development (evobrutinib, fenebrutinib, tolebrutinib) to penetrate the blood-brain barrier.
- To achieve this goal, we used *in silico* assessment based on Lipinski's Rule of Five (see table), *in vitro* kinase assays, cellular assays of apparent potency, and pharmacokinetic assessment of exposure of these candidates in plasma and CSF of the non-human primate cynomolgus (Macaca fascicularis) to predict the rank order of potency to inhibit BTK signalling in CNS-resident cells.

Generic Name	MW	cLogP	LogD (pH 7.4)	H-Donors	PSA	pKa	CNS MPO
Tolebrutinib	455.5	2.69	2.53	1	92.0	6.55	4.73
Evobrutinib	429.5	3.19	3.19	2	93.4	6.42	4.20
Ibrutinib	440.5	2.92	2.92	1	99.2	3.95	4.41
BIIB091	542.6	0.20	0.19	2	127.9	6.75	3.50
Remibrutinib	507.5	3.29	3.29	2	110.4	5.65	3.03
Fenebrutinib	664.8	1.59	1.57	2	119.3	7.55	3.52

METHODS

- **In silico prediction of CNS penetrance:** Multiparameter optimization (MPO) scores were derived from computationally accessible physicochemical descriptors All values for clogP, logD (pH 7.4), and polar surface area (PSA) were computed using ACDlabs (version 12). The number of H-donors was obtained from RDKit (version 2021.09.1), while the pKa value for the most basic center was computed using MoKa (version 2.6.6).
- **Biochemical kinase assays:** BTK activity was assessed by a third-party vendor (Nanosyn, Santa Clara, CA) using an assay mixture containing defined concentrations of inhibitor, 16 μM ATP, along with artificial, fluorescently-labeled substrate (FAMGEEPLYWSFPAKKK-NH2). The reaction was initiated by adding BTK (0.5 nM), and the reaction was followed for 3 hrs. at room temperature. A 12-point concentration response curve was constructed using the standard BTK kinase assay conditions. Phosphotransferase activity was monitored optically using a microfluidic Caliper system. Inhibitors were provided to the vendor in a blinded fashion, diluted in 100% DMSO using serial 3-fold dilution steps. Final compound concentrations in the assay ranged from 10 μM to 0.0565 nM. Compounds were tested in a single well for each dilution, and the final concentration of DMSO was kept at 1%. Reference compound, staurosporine, was tested in an identical manner.
- **Cellular activation assays:** Ramos cell (an immortalized human B cell line) expression of CD69 was used to evaluate cellular activity linked to BTK signaling. Defined concentrations of inhibitor were obtained via twelve serial 3-fold dilution steps ranging between 6 μM and 33.8 pM. B cell receptor was stimulated by adding goat α-Human IgM F(ab')2 at a final concentration of 20 μg/ml. The activated Ramos cells were incubated overnight (18 h) in 5% CO_2 at 37°C followed by staining for flow cytometry (BD LSR2) and analysis (FlowJo and Graphpad Prism).
- **Non-Human Primate (NHP) PK Studies:** Three healthy male animals were used in a crossover study conducted by a third-party vendor (Biomere, Worcester, MA). On Days 1-4, animals received a single daily oral dose of the test article at 10 mg/kg. The dose volume was 5 mL/kg. Following a 7-day washout period, the same three animals received a single daily oral dose of a second test article at 10 mg/kg. This sequence was repeated a third time for the final test article. Animal health checks were performed at least twice daily, in which all animals were checked for general health. Body weights were recorded prior to dosing on Day -1 or Day 1. CSF samples were collected from an indwelling intrathecal catheter accessed via subcutaneous port using sterile technique. Approximately 180 μL of fluid (saline lock) was removed prior to CSF sample collection at 1-, 2-, 4- and 8 hours post administration. All CSF samples were stored at -80ºC prior to analysis. Blood samples (~1 mL) were collected from an appropriate peripheral vein at 0.5-, 1-, 2-, 4-, 8-, and 24-hours post administration. Whole blood was collected into K_2EDTA tubes and placed on wet ice until processed to plasma, then stored at -80ºC for analysis.

RESULTS

Figure 1. Rank Order of Apparent Potency of BTK Inhibitors

	BTK			
	IC50 (nM)	Conf. Interval - 95%	Hill	Curve
Tolebrutinib	**0.676**	0.355	1.25	
Fenebrutinib	**6.21**	9.25	0.69	
Orelabrutinib	**14.7**	22.6	0.78	
Evobrutinib	**33.5**	44.4	0.88	

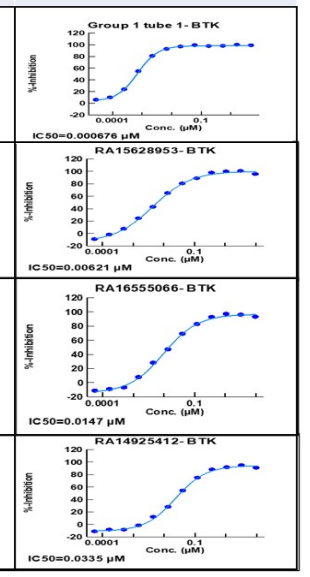

Group 1 tube 1- BTK
IC50=0.000676 µM
RA15628953- BTK
IC50=0.00621 µM
RA16555066- BTK
IC50=0.0147 µM
RA14925412- BTK
IC50=0.0335 µM

Figure 3. Tolebrutinib Inhibits B cell Activation with Nanomolar Potency

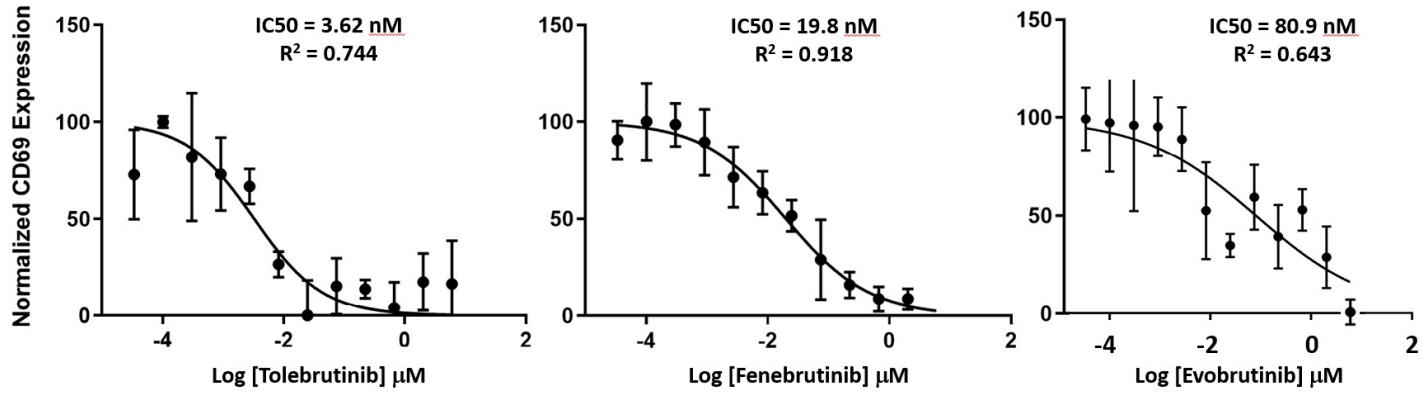

Normalized CD69 Expression
IC50 = 3.62 nM
R² = 0.744
Log [Tolebrutinib] µM
IC50 = 19.8 nM
R² = 0.918
Log [Fenebrutinib] µM
IC50 = 80.9 nM
R² = 0.643
Log [Evobrutinib] µM

Figure 5. Partition Coefficient Confirms Tolebrutinib Intrinsic CNS Penetrance

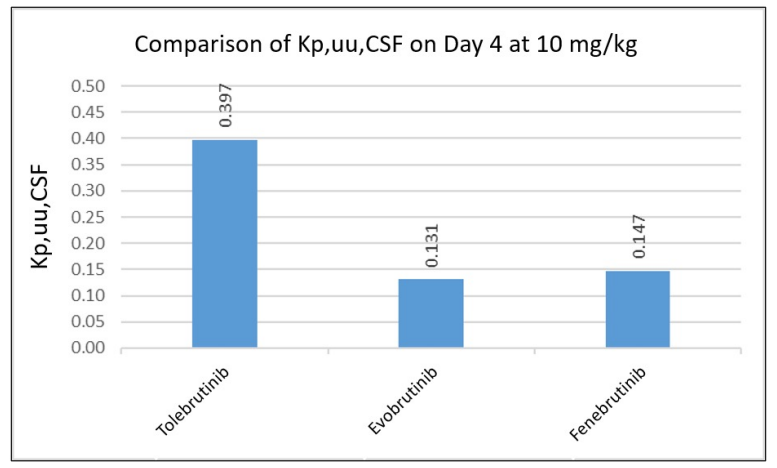

Comparison of Kp,uu,CSF on Day 4 at 10 mg/kg
Kp,uu,CSF
0.397
0.131
0.147
Tolebrutinib
Evobrutinib
Fenebrutinib

BTK Inhibitor	AUC, CSF (h*ng/mL)	Cmax, CSF (ng/mL)
Tolebrutinib	19.6	4.84
Evobrutinib	10.8	3.20
Fenebrutinib	62.3	12.9

Figure 2. Tolebrutinib inhibited BTK activity 64x faster than evobrutinib, 1780x faster than fenebrutinib

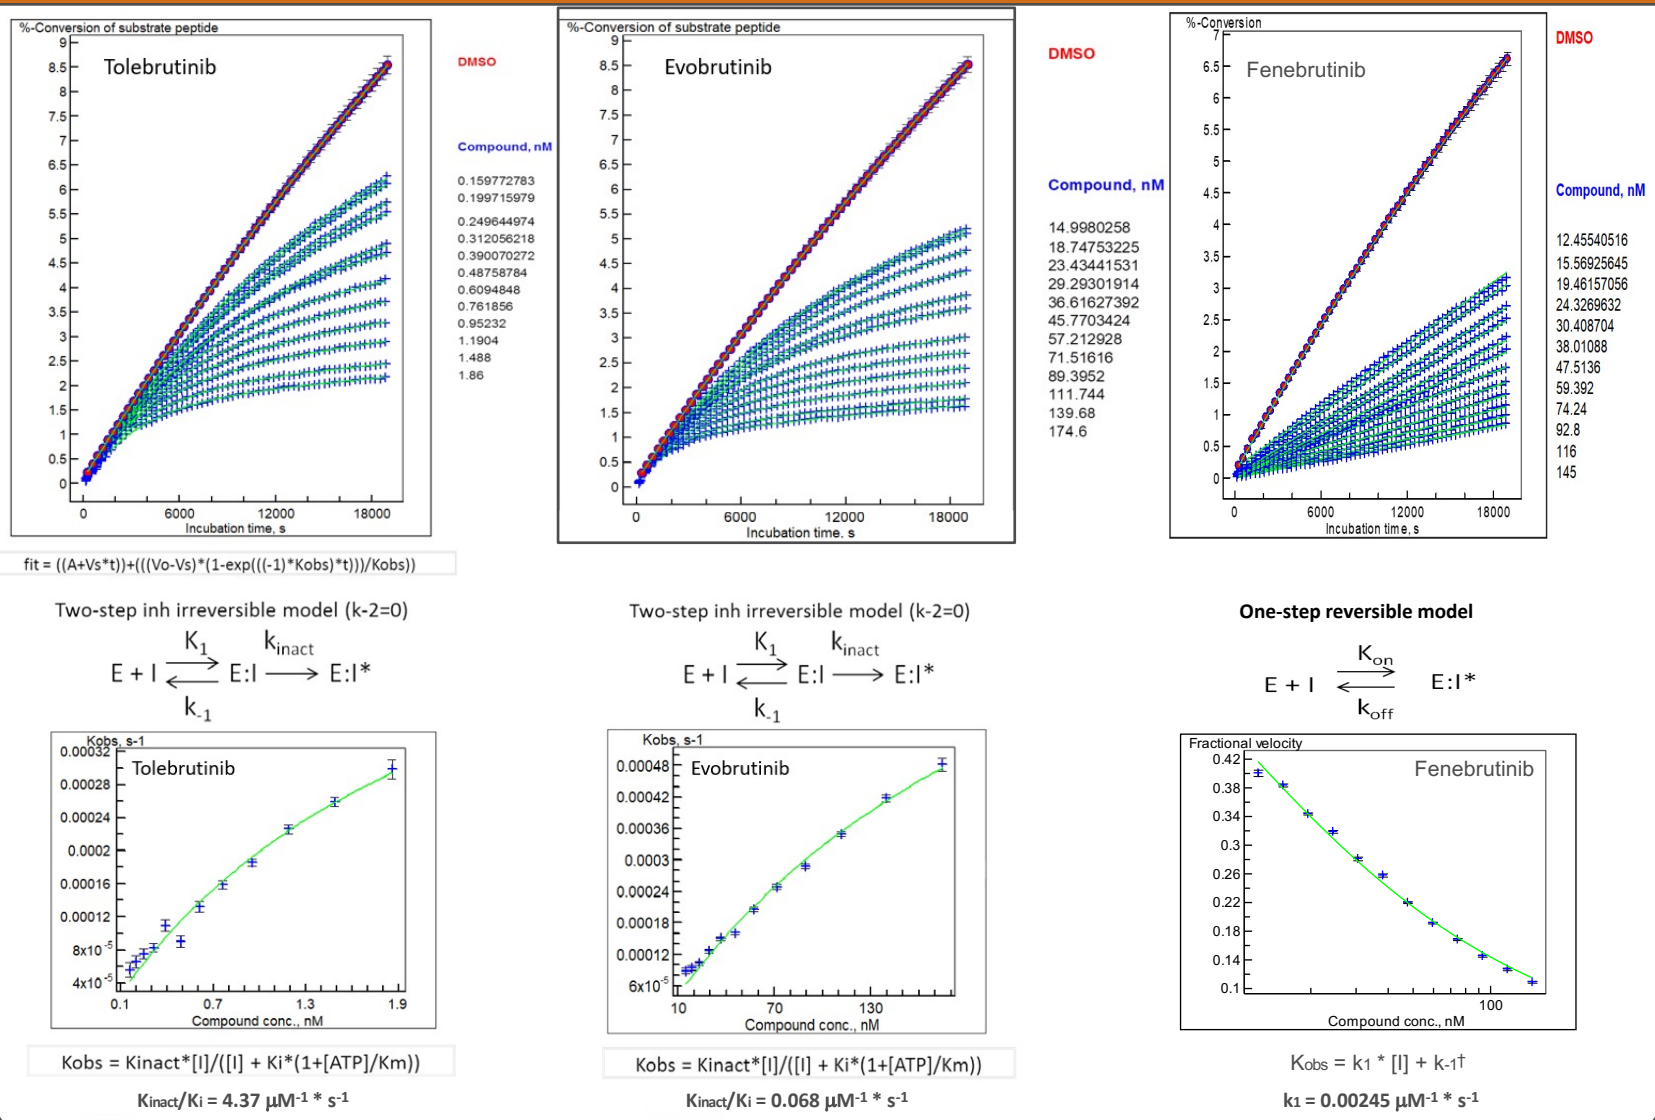

%-Conversion of substrate peptide
Tolebrutinib
DMSO
Compound, nM
0.159772783
0.199715979
0.249644974
0.312056218
0.390070272
0.48758784
0.6094848
0.761856
0.95232
1.1904
1.488
1.86
Incubation time, s
fit = ((A+Vs*t))+(((Vo-Vs)*(1-exp(((-1)*Kobs)*t)))/Kobs))
Two-step inh irreversible model (k-2=0)
Kobs, s-1
Compound conc., nM
Kobs = Kinact*[I]/([I] + Ki*(1+[ATP]/Km))
Kinact/Ki = 4.37 µM-1 * s-1
Evobrutinib
DMSO
Compound, nM
14.9980258
18.74753225
23.43441531
29.29301914
36.61627392
45.7703424
57.212928
71.51616
89.3952
111.744
139.68
174.6
Two-step inh irreversible model (k-2=0)
Kobs = Kinact*[I]/([I] + Ki*(1+[ATP]/Km))
Kinact/Ki = 0.068 µM-1 * s-1
%-Conversion
Fenebrutinib
DMSO
Compound, nM
12.45540516
15.56925645
19.46157056
24.3269632
30.408704
38.01088
47.5136
59.392
74.24
92.8
116
145
One-step reversible model
Fractional velocity
Kobs = k1 * [I] + k-1†
k1 = 0.00245 µM-1 * s-1

† Value for k_{-1} (1.52×10^{-5} s^{-1}) from Crawford (2018)

Figure 4. CSF Exposure in NHP Exceeded the IC90 for Tolebrutinib

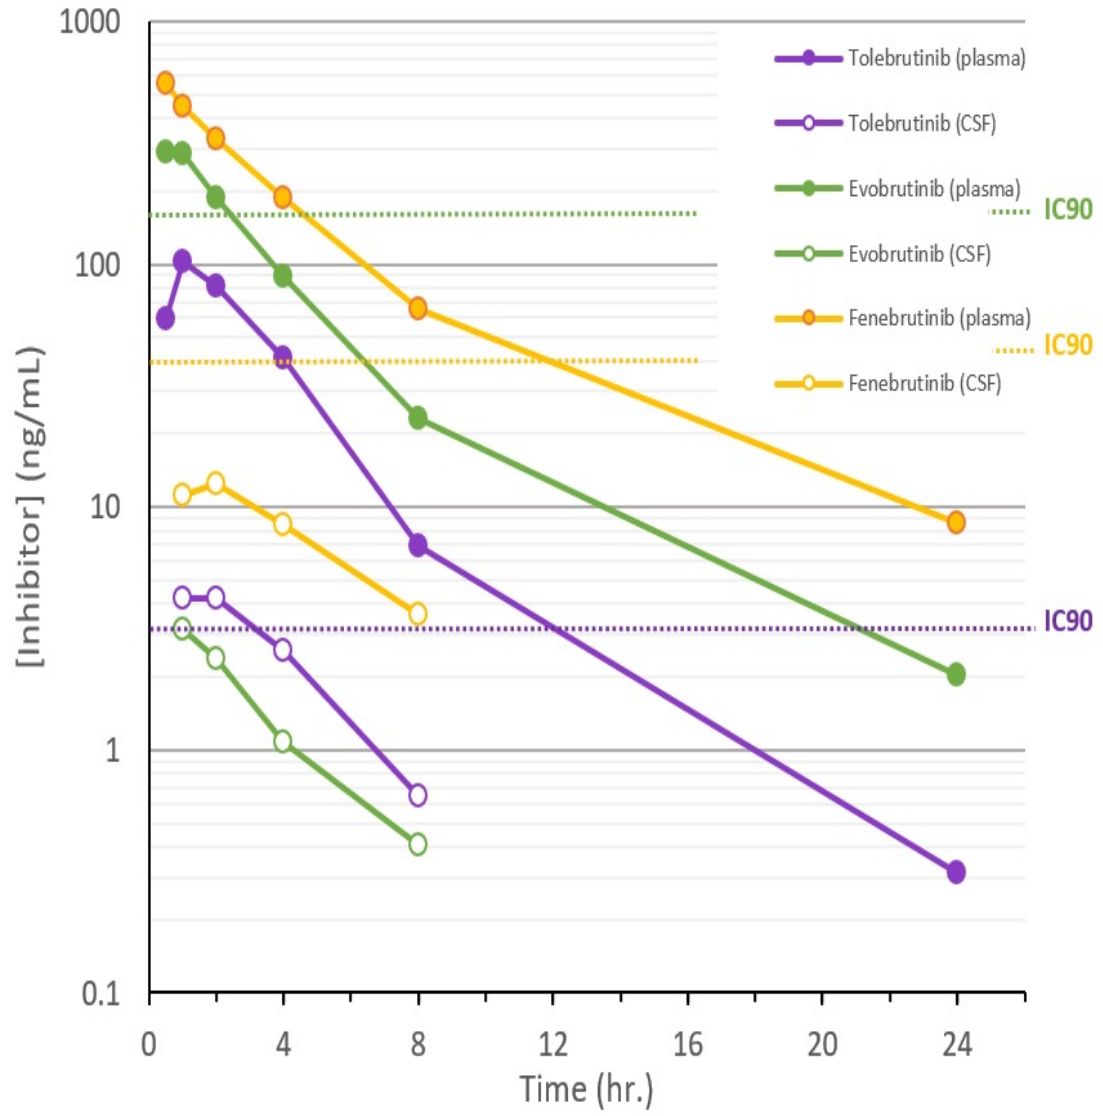

[Inhibitor] (ng/mL)
Time (hr.)
Tolebrutinib (plasma)
Tolebrutinib (CSF)
Evobrutinib (plasma)
Evobrutinib (CSF)
Fenebrutinib (plasma)
Fenebrutinib (CSF)
IC90
IC90
IC90

CONCLUSIONS

- **Tolebrutinib was more potent than evobrutinib (50x) or fenebrutinib (9.3x)**
- **Tolebrutinib inhibited BTK 64x faster than evobrutinib, 1780x faster than fenebrutinib**
- **Relative potency to inhibit Ramos cell activation was consistent with biochemical results**
- **Target engagement by the irreversible inhibitors was driven by AUC, not C_{max}**
- **The two irreversible inhibitors produce durable inhibition, while fenebrutinib is reversible and inhibition relies on sustained drug exposures**
- **The slow first-order rate constant for fenebrutinib indicated that target engagement was driven by C_{trough}**
- **Only tolebrutinib exceeded the IC90 value in the CNS of NHPs**
- **Tolebrutinib demonstrated intrinsic CNS penetrance based on the unbound partition coefficient Kp,uu = 0.397, roughly 3x higher than evobrutinib (0.131), fenebrutinib (0.147)**
- **The combination of high potency, reaction rates, and CNS exposure suggested that tolebrutinib inhibits BTK signalling in the CNS by >90%, consistent with pharmacological activity in the brain and spinal cord**

Acknowledgments and Disclosures

TT, PB, DO: Employees of Sanofi (may hold shares and/or stock options in the company); RCG: Employee of Sanofi at time of study, but current employee of Takeda.

Presented at the Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS) Forum, February 24–26, 2022, West Palm Beach, FL, USA

Funding provided by Sanofi